A Phase I Study of the First in Class Mitochondrial Metabolism Inhibitor CPI-613 in Patients with Advanced Hematologic Malignancies

Timothy S. Pardee MD, PhD, Denise Levitan MD, David Hurd MD, Leslie R. Ellis MD, Scott Isom MS, Robin Harrelson BSN, Megan Manuel MSN, Sarah Dralle MSN, Susan Lyerly PA-C and Bayard L. Powell MD

Department of Internal Medicine, Section on Hematology, Wake Forest School of Medicine, Winston-Salem, NC; [2]Department of Cancer Biology, Wake Forest School of Medicine, Winston-Salem, NC

Wake Forest® School of Medicine

CPI-613 is a Novel Lipoate Derivative

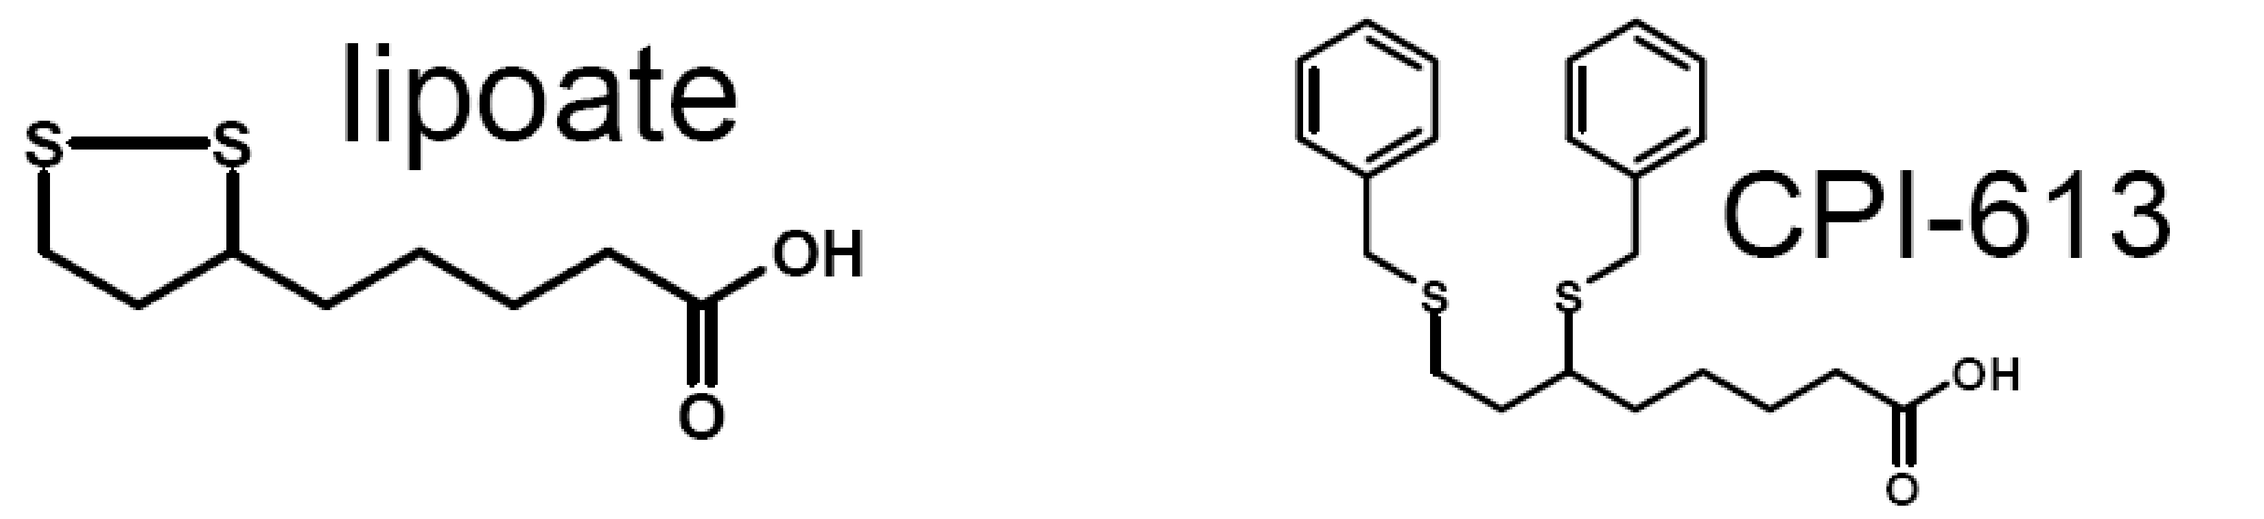
lipoate
CPI-613

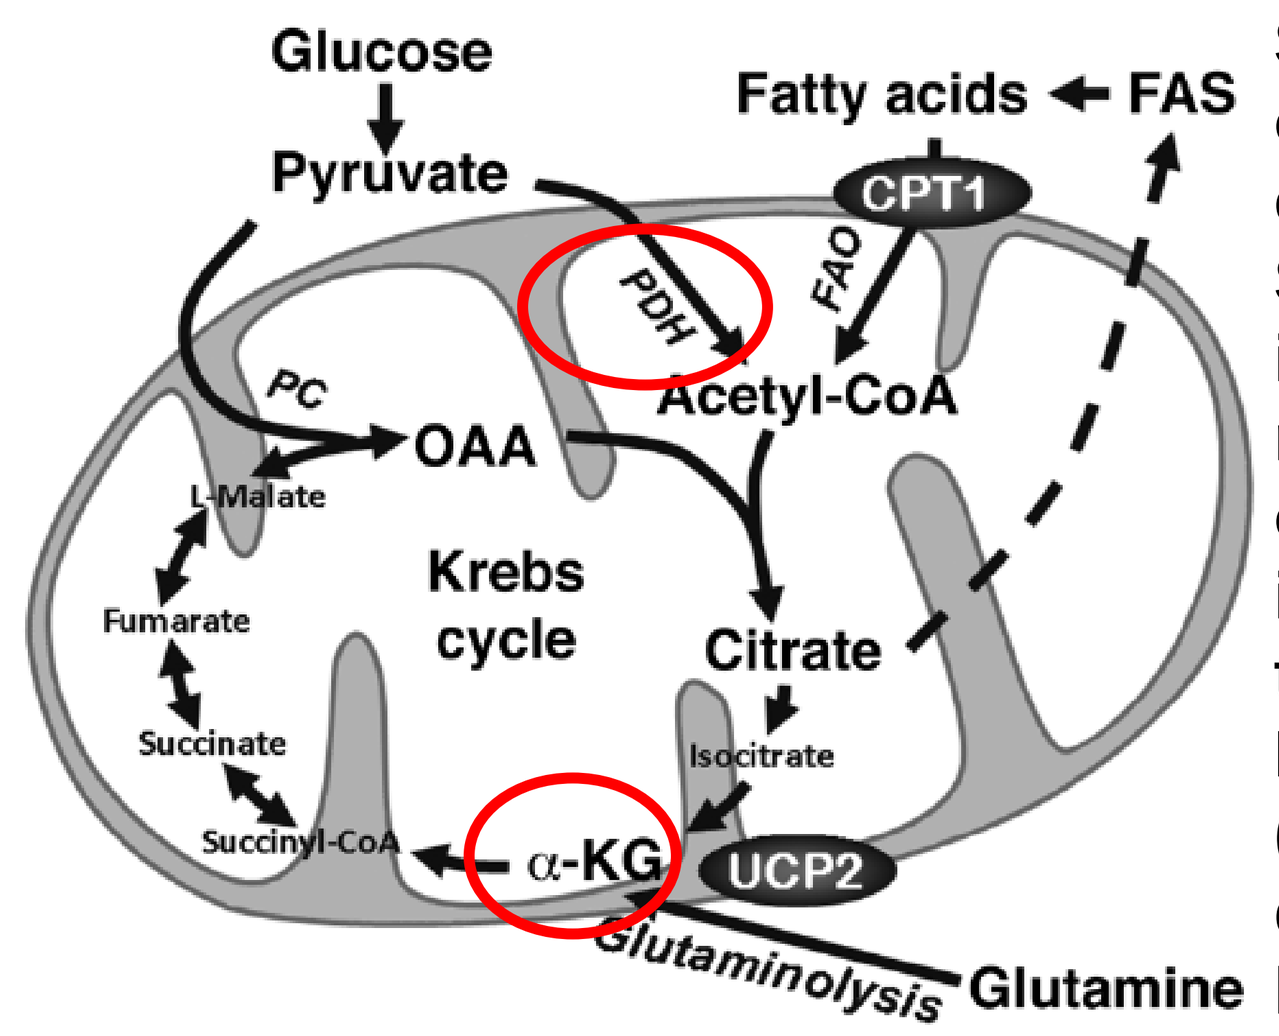
Glucose
Pyruvate
Fatty acids
FAS
CPT1
PDH
FAO
Acetyl-CoA
PC
OAA
L-Malate
Fumarate
Succinate
Succinyl-CoA
Krebs cycle
Citrate
Isocitrate
α-KG
UCP2
Glutaminolysis
Glutamine

Small molecules that inhibit a single oncogenic pathway have not proven efficacious in most malignancies. Strategies that target pathways active in all cancer cells regardless of driving mutations will likely improve outcomes. Mitochondrial metabolism is widely aberrant in cancer including the hematologic malignancies. Pyruvate dehydrogenase complex (PDH) and α-Ketogluterate dehydrogenase complex (α-KG) are 2 key mitochondrial enzymes that require lipoate for their function. CPI-613 is a non-redox active lipoate derivative that inhibits these enzymes developed by Cornerstone Pharmaceuticals.

Samudio, et. al., *J Clin Invest*. 2010;120(1):142-56

Zachar et. al. *J Mol Med*. 2011 Nov;89(11):1137-48

CPI-613 is Selectively Taken up by Transformed Cells

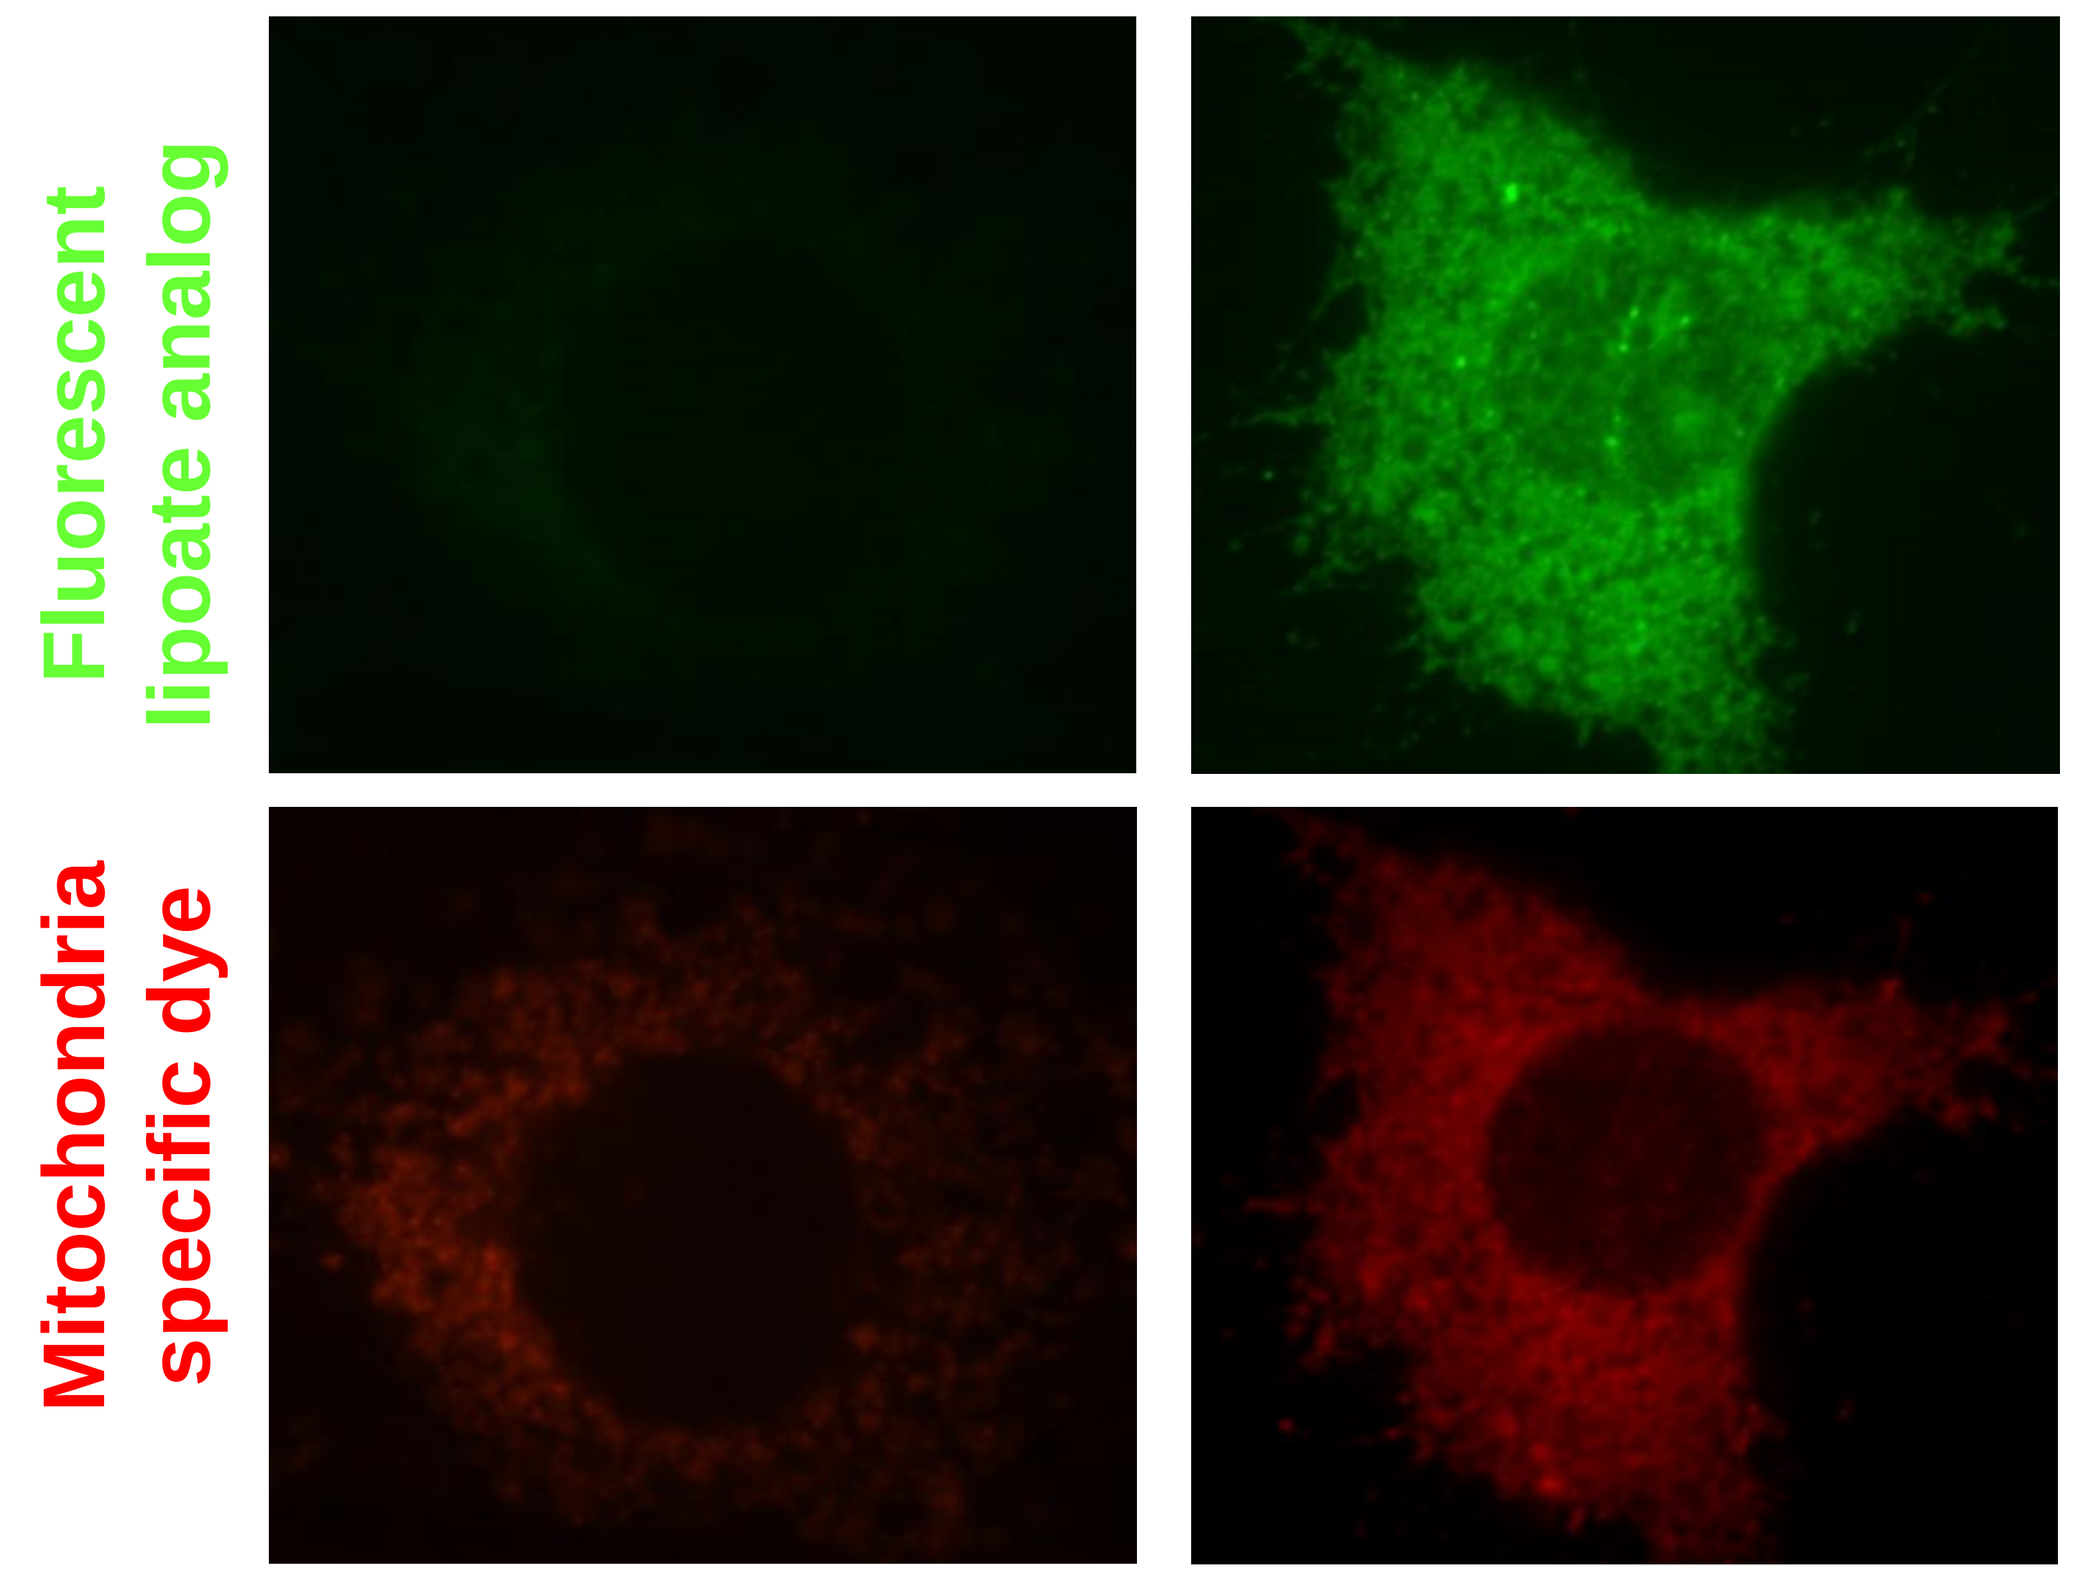
NIH 3T3
Ras, NIH 3T3
Fluorescent lipoate analog
Mitochondria specific dye

CORNERSTONE PHARMACEUTICALS

Stuart, Zachar & Bingham, unpublished

Phase I Clinical Trial for Patients with Advanced Heme Malignancies

Schema:

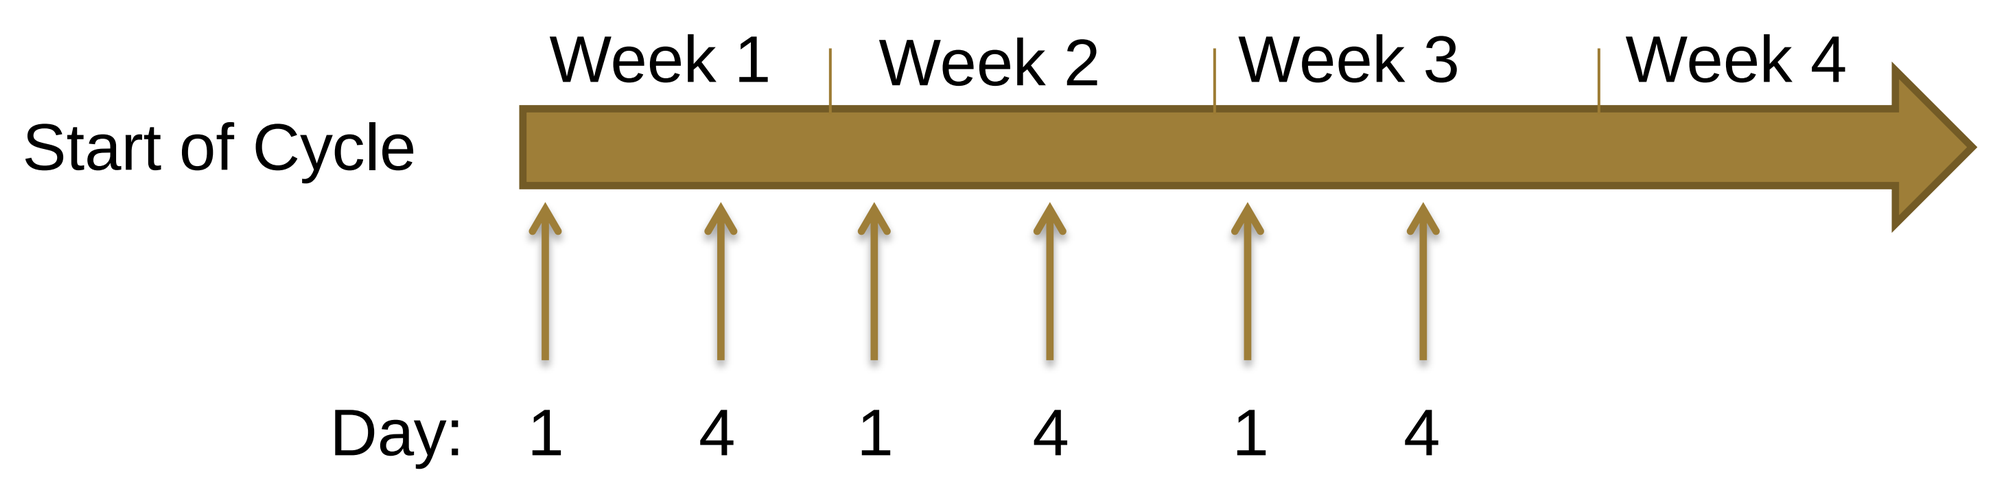
Start of Cycle
Week 1
Week 2
Week 3
Week 4
Day: 1 4 1 4 1 4

This was a Phase I open label trial using a 2-stage dose-escalation scheme (single-patient & traditional stages):
Single-Patient Dose-Escalation Stage: In the single-patient stage, a single patient was accrued per dose level. The starting dose was 420 mg/m². Dose level was escalated (by doubling the previous dose) until toxicity >Grade 1, then the traditional dose-escalation was triggered.
Traditional Dose-Escalation: All dose escalations are according to the modified Fibonacci Dose-Escalation scheme. The dose level for the first cohort in the Traditional Dose-Escalation was 840 mg/m². The MTD was established at 2940 mg/m².
Treatment Cycle and Duration of Treatment: A treatment cycle is 3 consecutive weeks of 2x weekly, 2 hour infusions of CPI-613, followed by 1 week of rest. Patients were continued on trial if they appeared to have clinical benefit as determined by the treating physician.

CPI-613 has Activity in Burkitt's Lymphoma, Patient #14

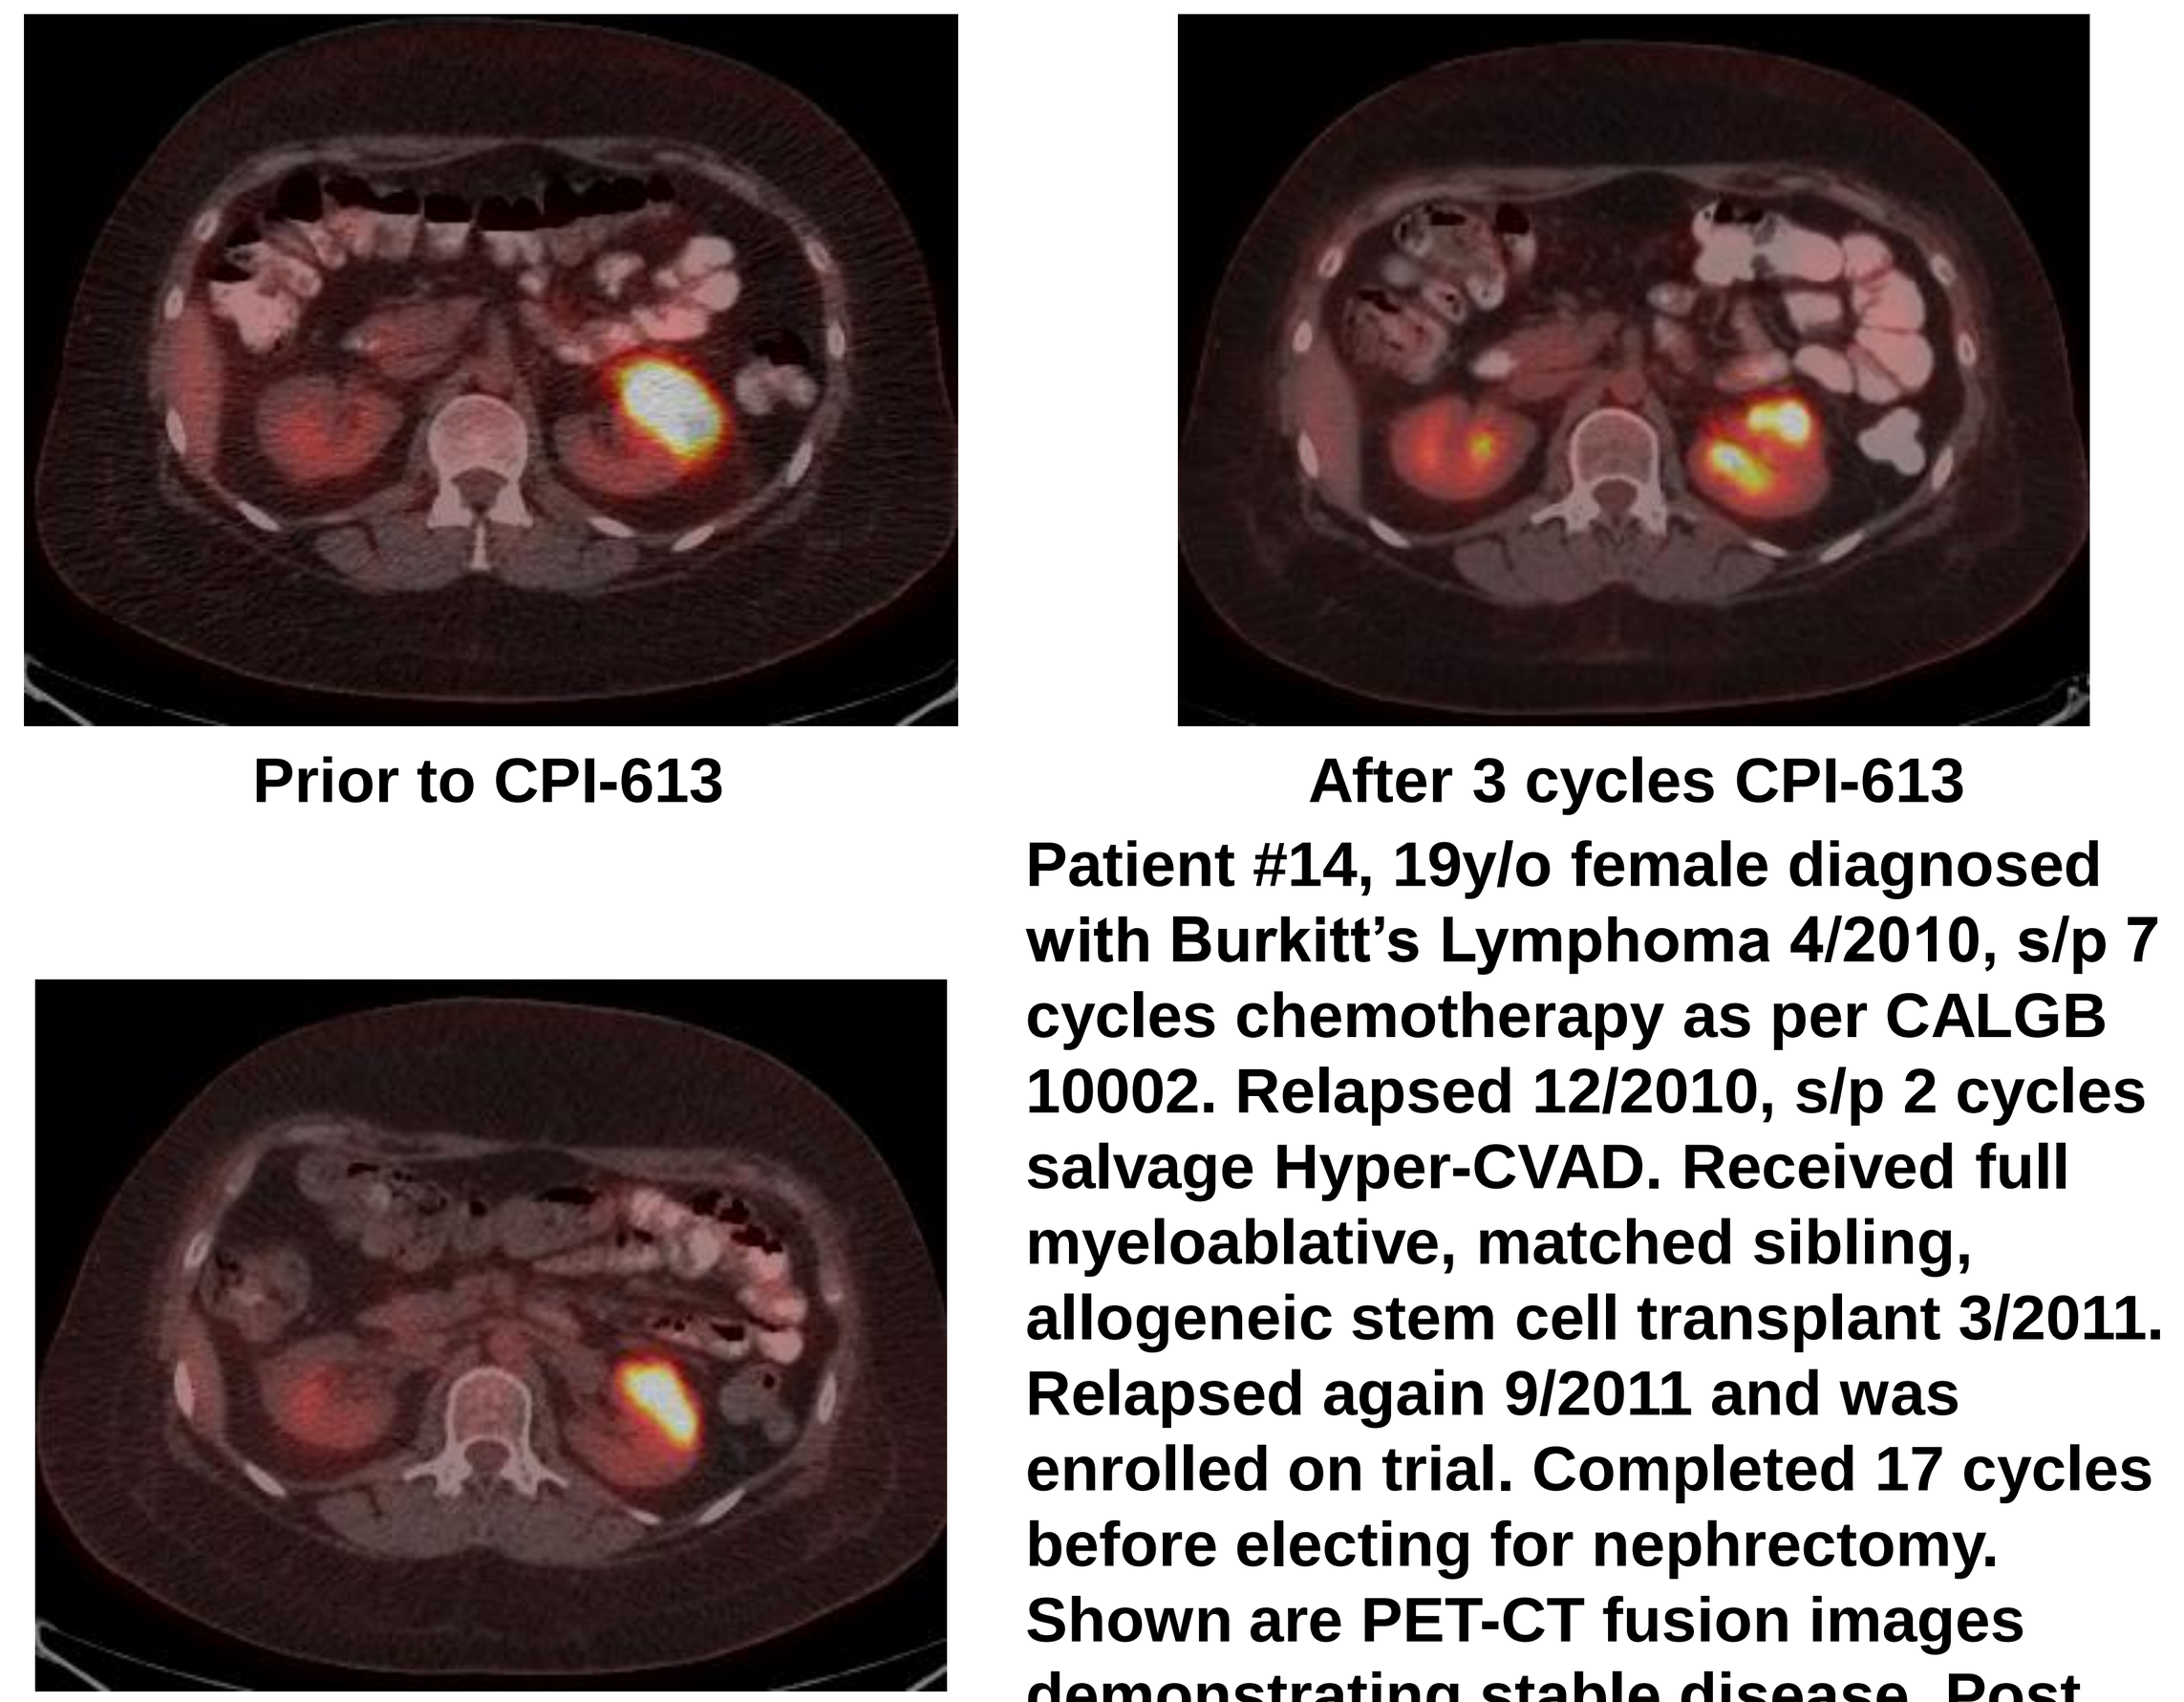
Prior to CPI-613
After 3 cycles CPI-613
After 5 cycles CPI-613

Patient #14, 19y/o female diagnosed with Burkitt's Lymphoma 4/2010, s/p 7 cycles chemotherapy as per CALGB 10002. Relapsed 12/2010, s/p 2 cycles salvage Hyper-CVAD. Received full myeloablative, matched sibling, allogeneic stem cell transplant 3/2011. Relapsed again 9/2011 and was enrolled on trial. Completed 17 cycles before electing for nephrectomy. Shown are PET-CT fusion images demonstrating stable disease. Post nephrectomy PET-CT scan revealed no evidence of disease.

CPI-613 has Activity in MDS, Patient #4

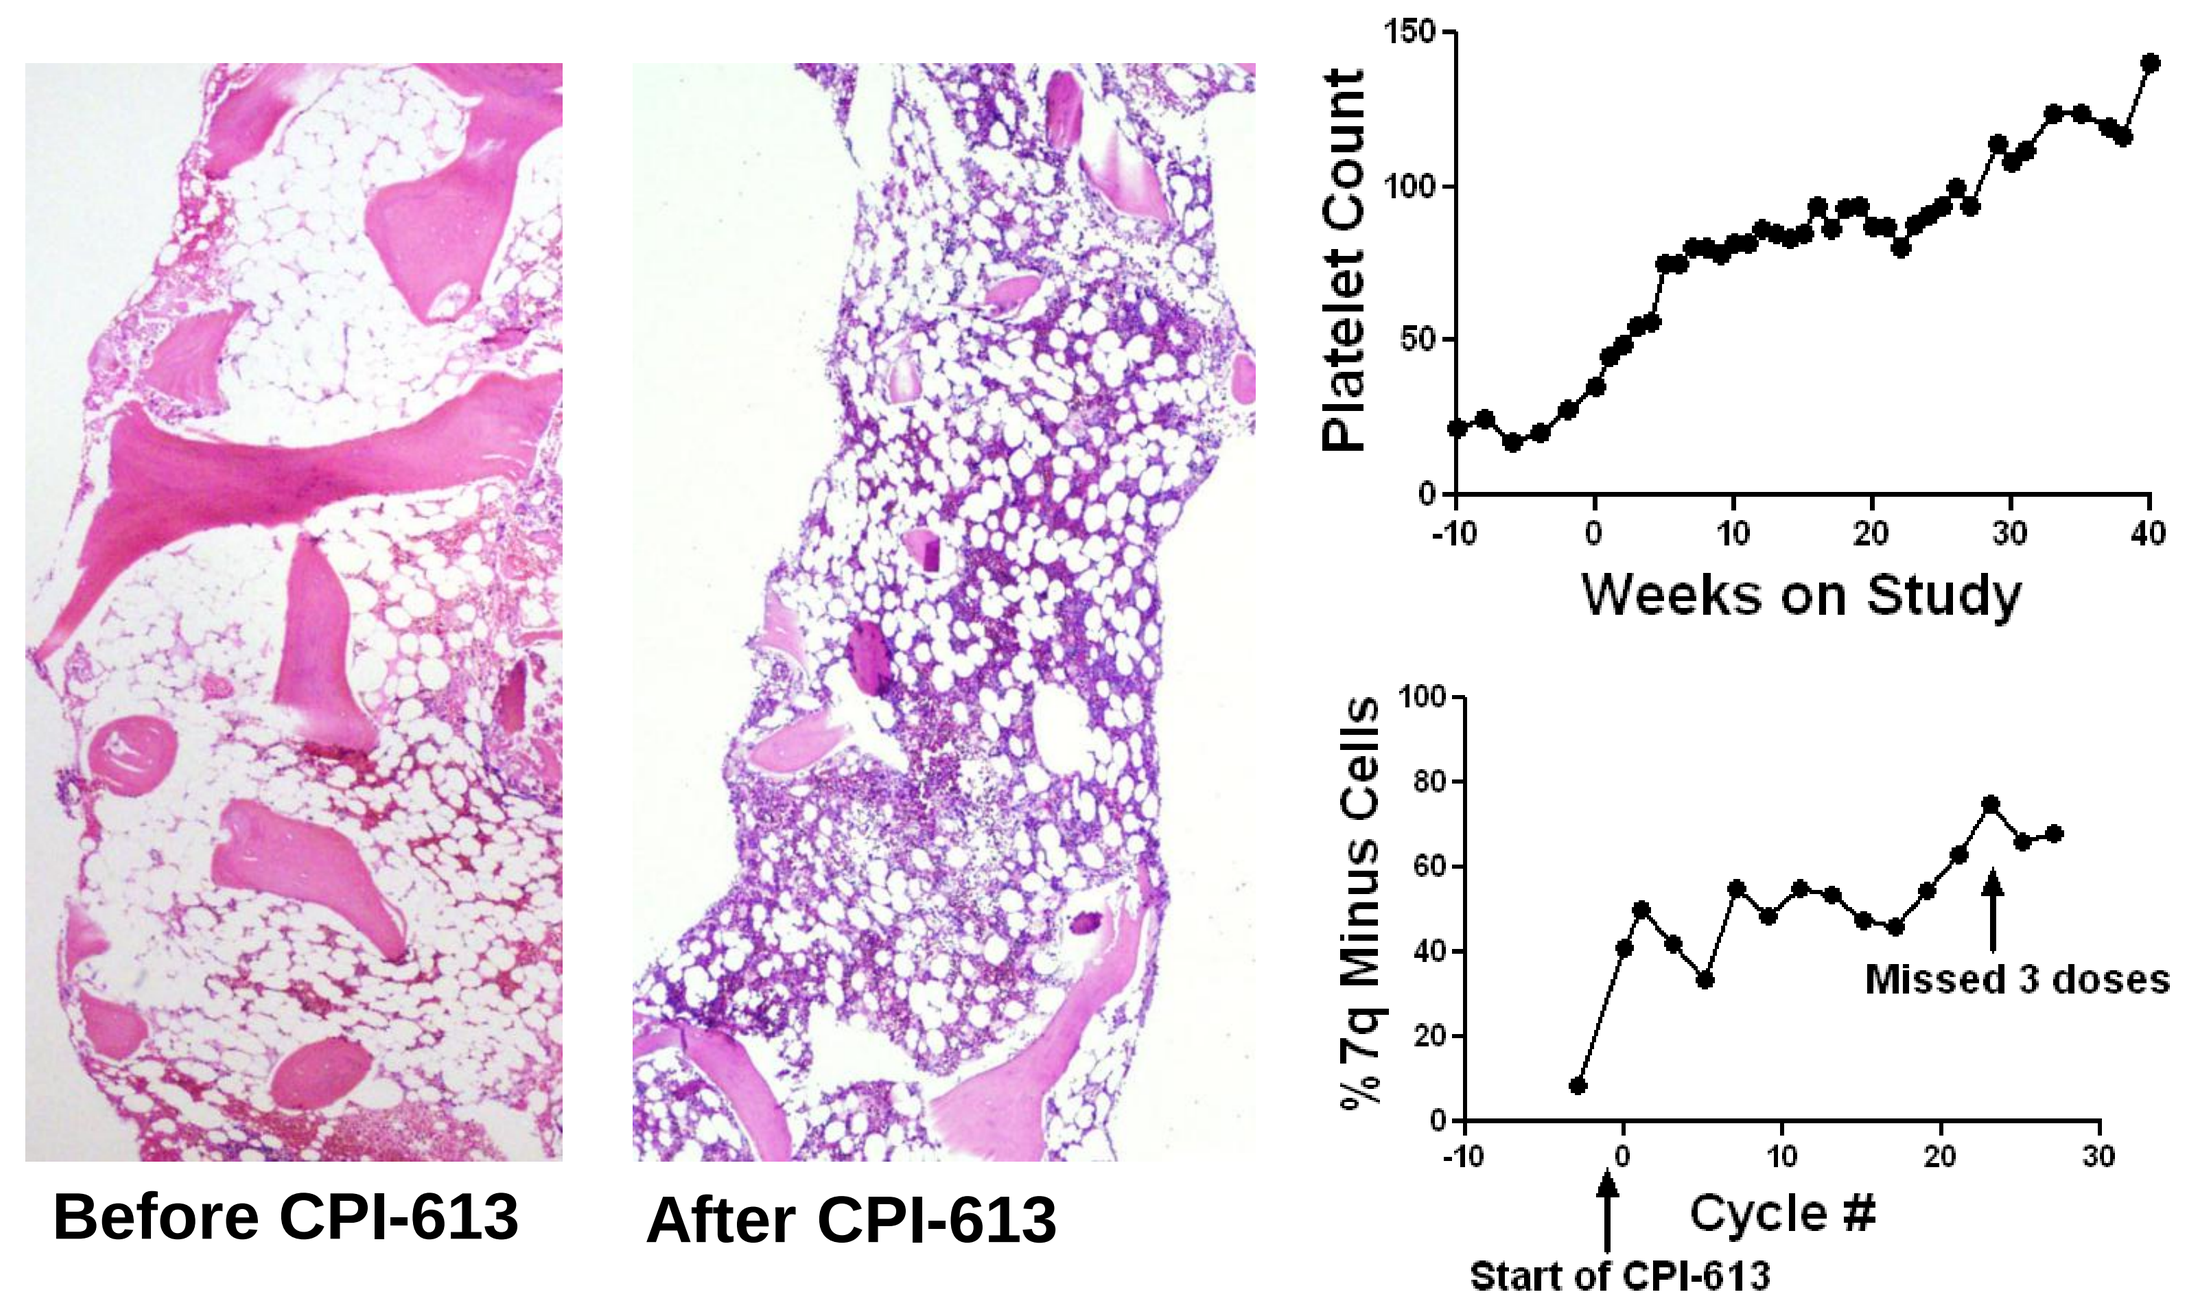
Before CPI-613
After CPI-613
Platelet Count
Weeks on Study
% 7q Minus Cells
Missed 3 doses
Cycle #
Start of CPI-613

Patient #4, a 49 y/o male diagnosed with AML with normal Karyotype in 2004. After standard therapy achieved CR1. Relapsed in 2008, received a HDAC based regimen and achieved CR2. Underwent autologous BMT in 2010 while in CR2. Delayed count recovery, red cell and platelet transfusion dependent. Three months post transplant found to have 8.5% cells with 7q- consistent with therapy related MDS. Marrow 6 months post transplant showing 41% cells with 7q-, remained transfusion dependent. Patient has been transfusion independent since starting CPI-613 and now is in CR3. He has been on continuous CPI-613 therapy for 31 cycles and remains in continuos remission.

CPI-613 has Activity in AML, Patient #12

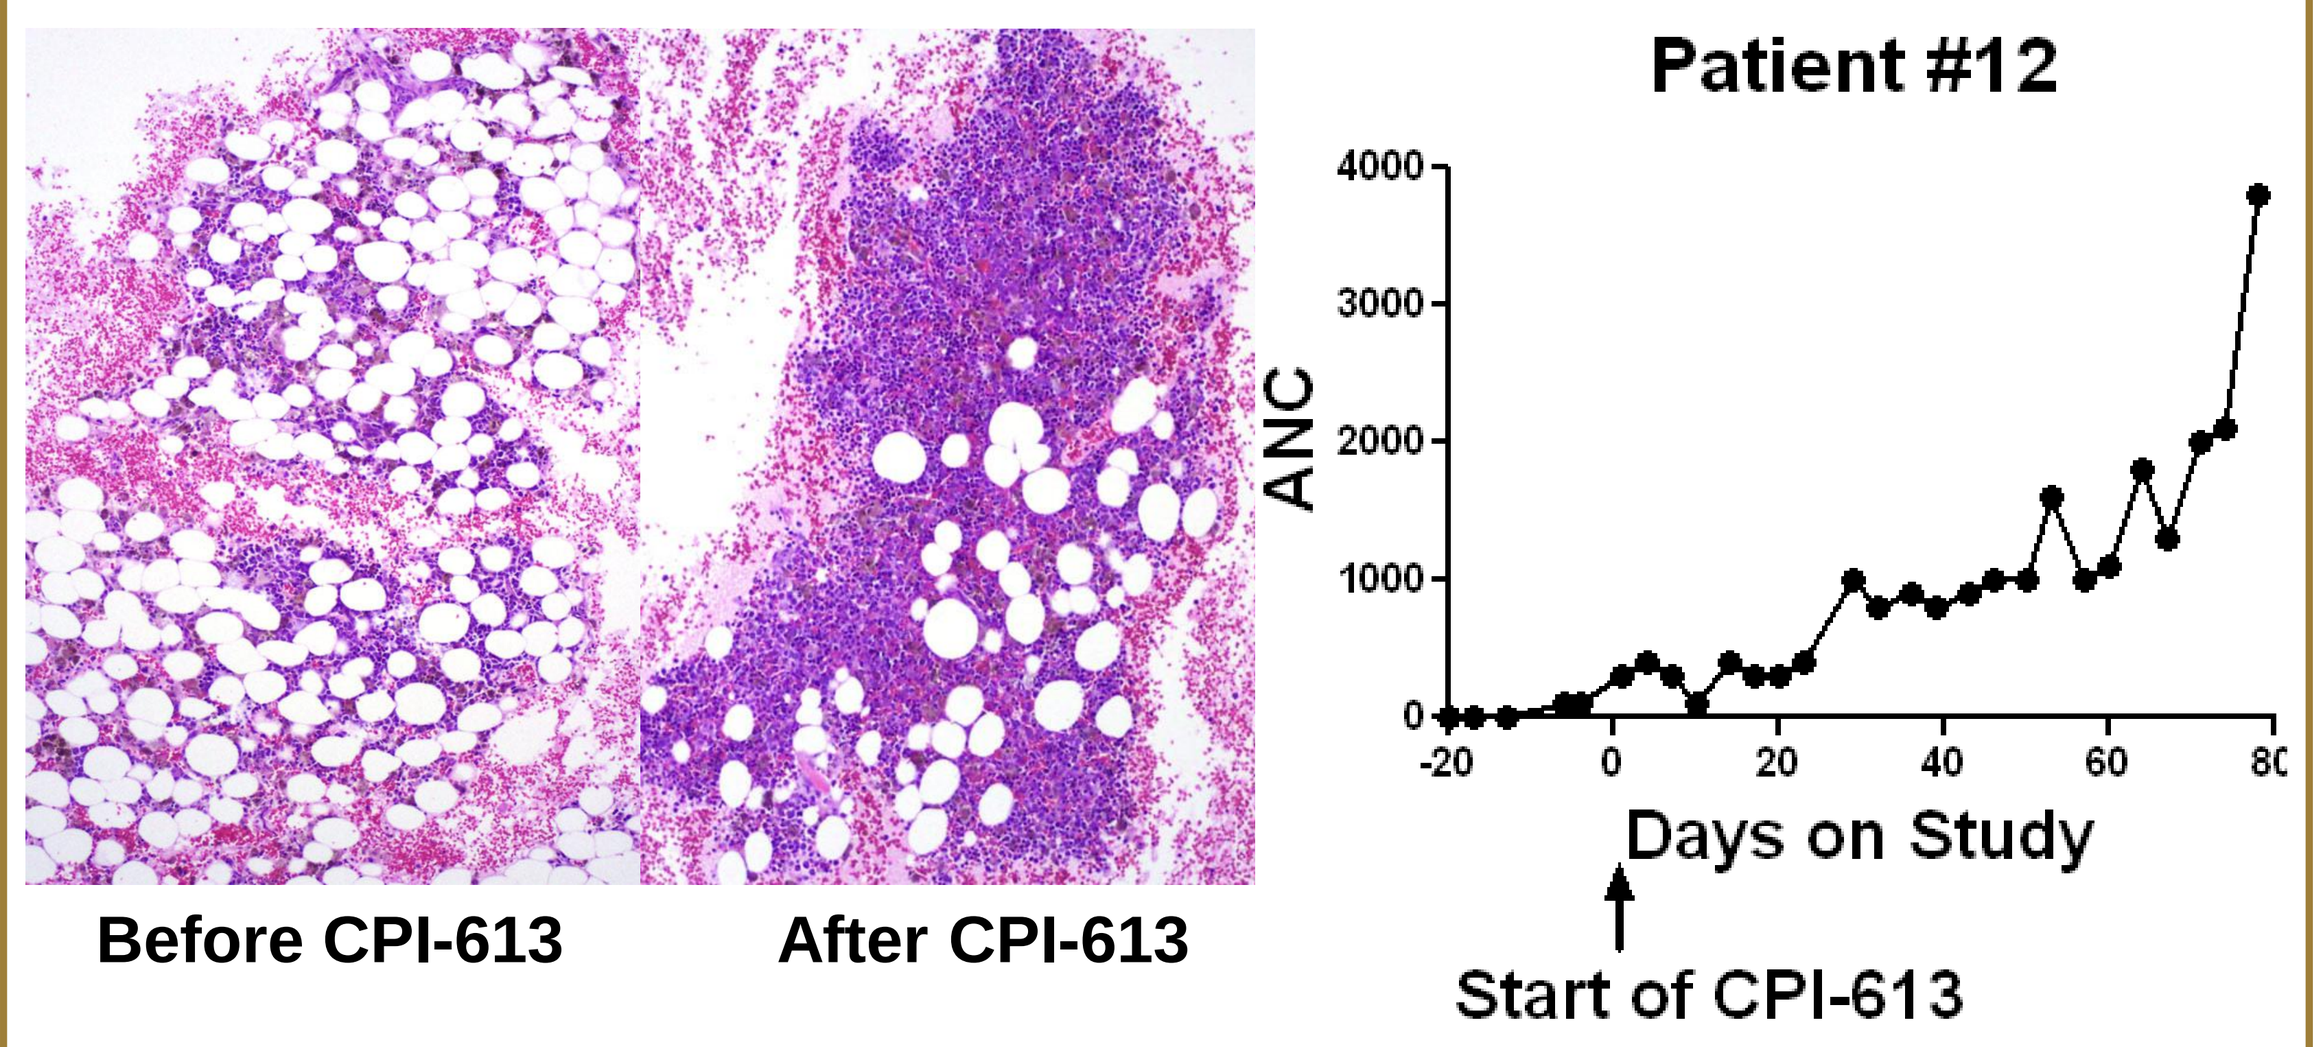
Before CPI-613
After CPI-613
Patient #12
ANC
Days on Study
Start of CPI-613

Patient #12, 66 y/o female with refractory AML. She received induction therapy with 7+3, 1st salvage attempt with cytoxan-etoposide, next salvage with high dose cytarabine-mitoxantrone-L-spar, then decitabine and finally azacytidine before starting on CPI-613. Pre-trial bone marrow biopsy showed hypocellular marrow with 9% blasts and 22% immature monocytes. After 2 cycles repeat biopsy shows hypercellular marrow with no evidence of disease. She was removed from study to undergo reduced intensity conditioned allogeneic stem cell transplant. Shown top left marrow clot section before and after 2 cycles CPI613, top right absolute neutrophil count (ANC).

CPI-613 has Activity in CTCL, Patient #20

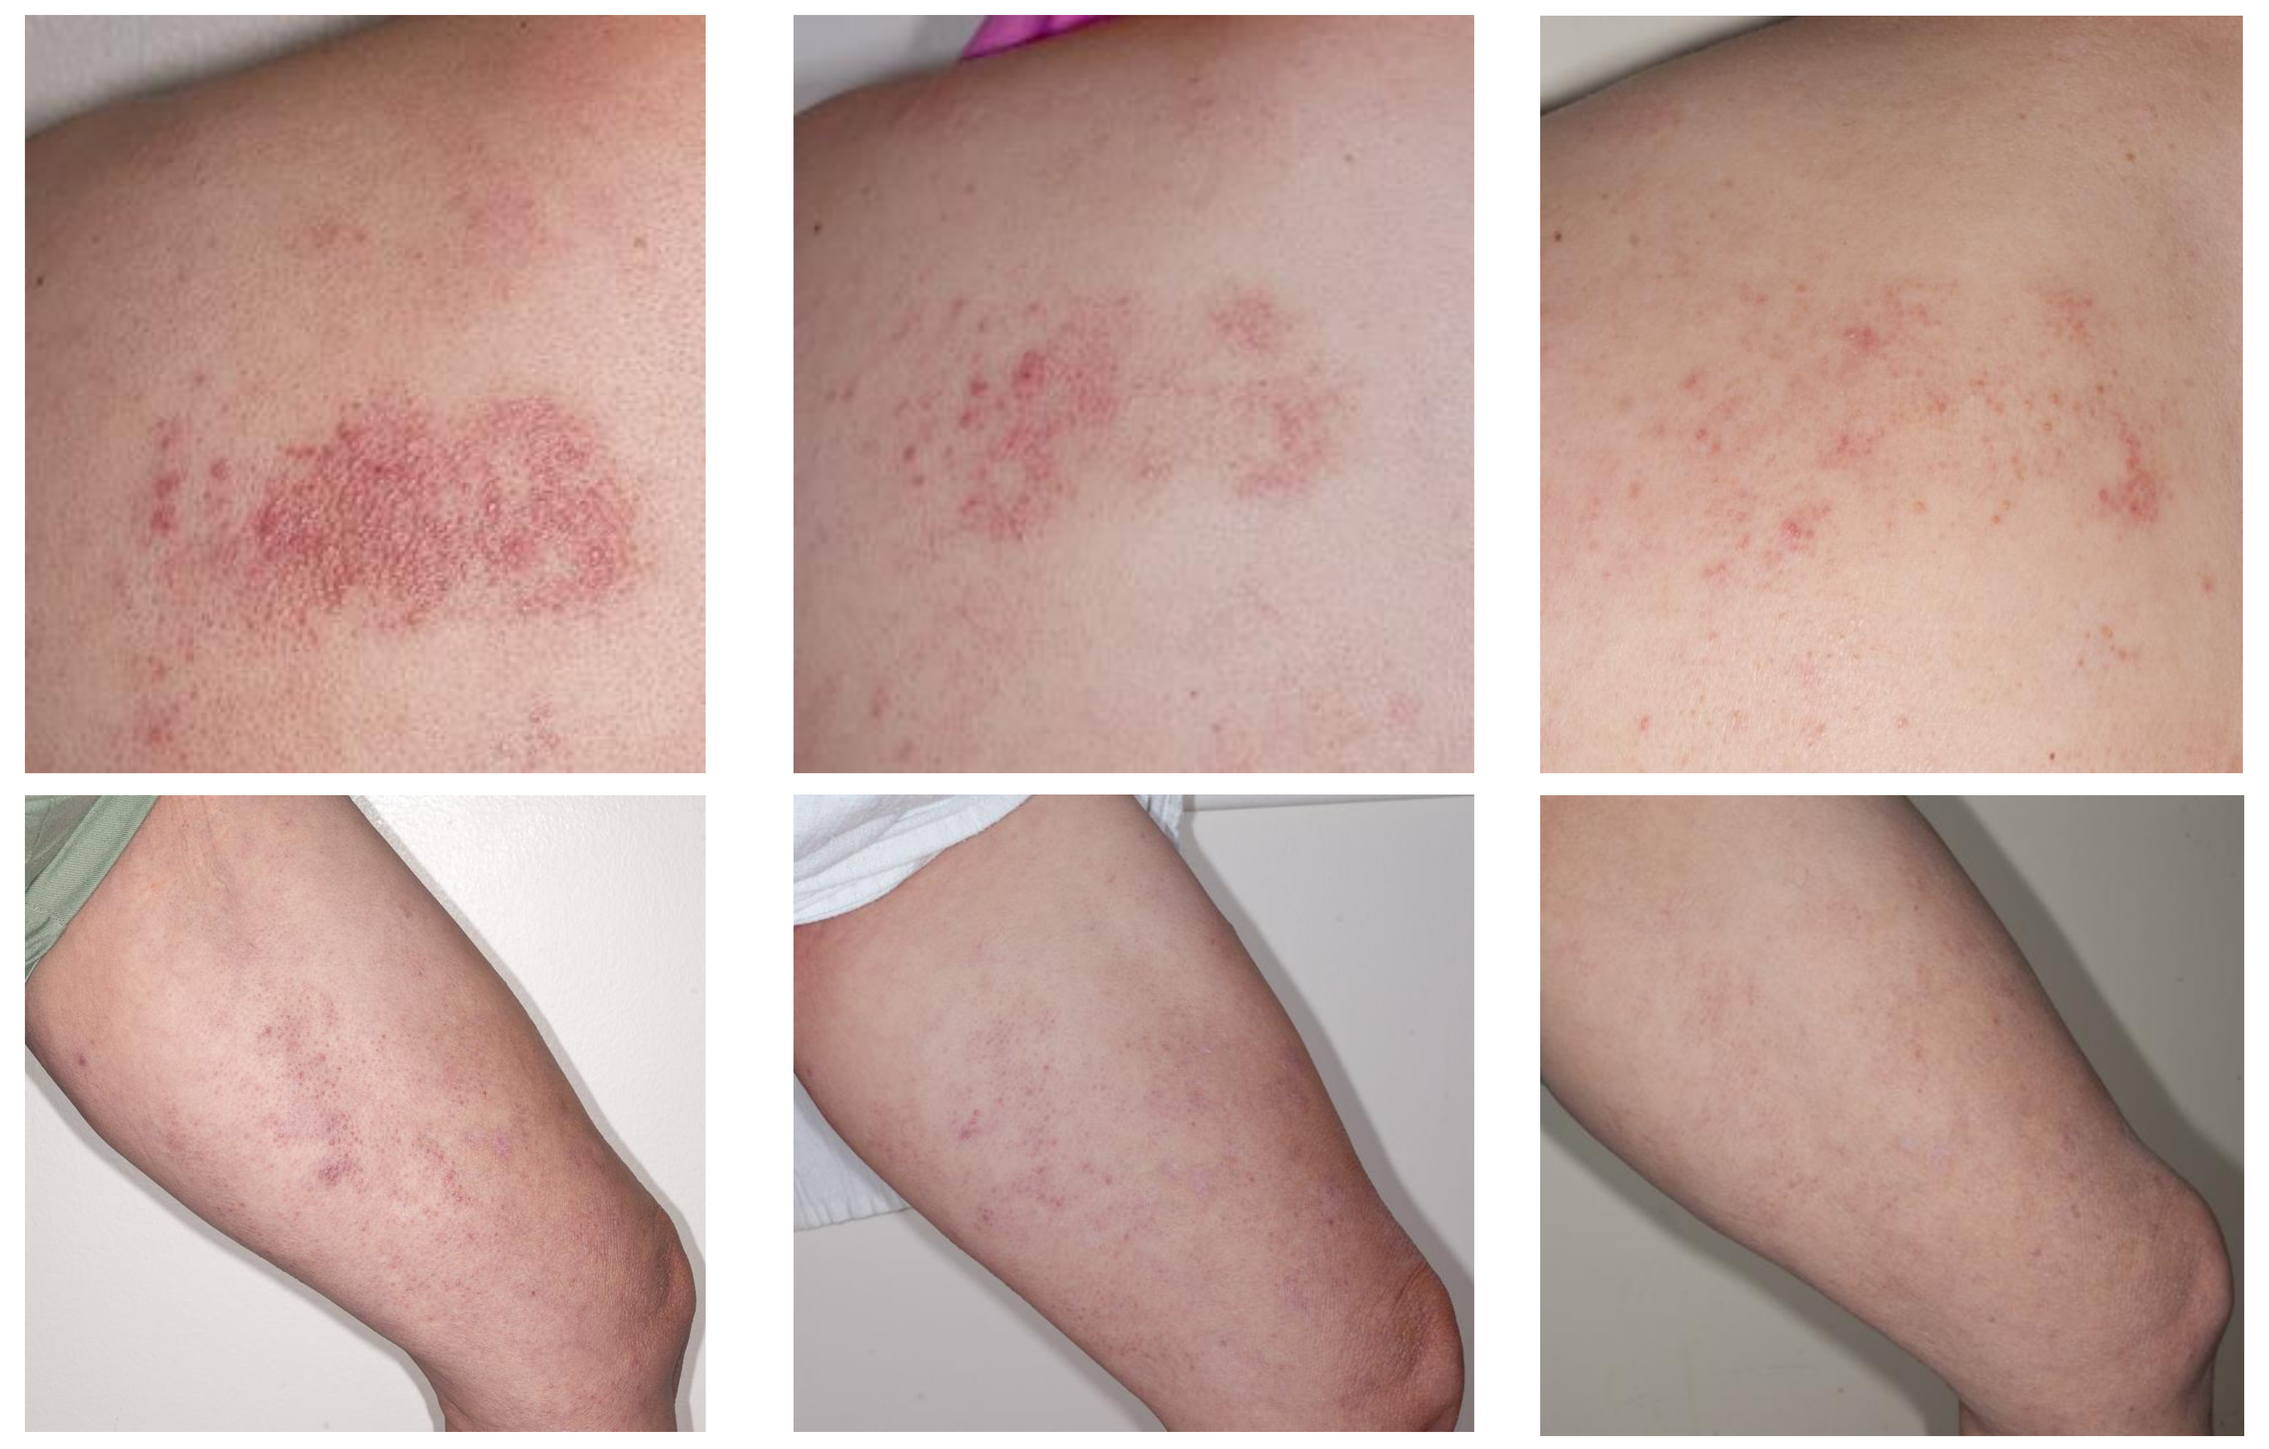
Cycle 1 Day 22
Cycle 4 Day 22
Cycle 7 Day 22

Patient #20, a 50 y/o female diagnosed with cutaneous T cell lymphoma (CTCL) in 6/2008. She was treated with oral methotrexate, Targretin, Zolinza, Ontak, Gemcitabine, Pralatrexate, Romidpesin, Velcade, Romidpesin, and Zolinza prior to her enrollment on trial. Started CPI-613 as 11th line therapy and has achieved a PR. She petitioned to have her week of rest removed secondary to flares of her disease during the off week and has remained on continuos therapy for 13 cycles.

CPI-613 is Well Tolerated

Attribution = Probable + Definite				
Toxicity	Grade 2	Grade 3	Grade 4	Grade 5
Nausea	1	0	0	0
Vomiting	0	1	0	0
Diarrhea	3	0	0	0
Creatinine (0.5-1.5)	0	3	0	0
Proteinuria (neg, trace)	1	0	0	0
Renal failure	0	3	0	0
Hypotension	1	0	0	0
Calcium, serum-low (hypocalcemia)	1	0	0	0
Albumin, serum-low (hypoalbuminemia)	1	0	0	0
Potassium, serum-high (hyperkalemia)	0	1	0	0
Leukopenia	0	0	0	0
Anemia	0	0	0	0
Thrombocytopenia	0	0	0	0
Glomerular filtration rate	0	2	0	0

DLTs were prolonged grade 3 nausea and acute renal failure at a dose of 3780 mg/m². No marrow suppression at any dose was attributed as probable or definitely associated with CPI-613.

CPI-613 Clinical Activity Summary

Patient #	Diagnosis	Dose mg/m²	Best response	> 1 cycle?
1	NHL	420	NA	No
2	AML	420	PD	No
3	Myeloma	840	PD	No
4	MDS	840 ->2100	CR	Yes (31 cycles)
5	Hodgkin's	840	PD	No
6	AML	1386	PD	No
7	Myeloma	1386	SD	Yes (3 cycles)
8	Myeloma	1386	SD	Yes (4 cycles)
9	AML	1386	PD	No
10	MDS (RAEB-2)	2100	SD	Yes (8 cycles)
11	MDS (RA)	2100 ->2940->1470	SD	Yes (12 cycles)
12	AML	2100	MLFS	Yes (2 cycles)
13	MDS	2940	NA	No
14	Burkitt's	2940	PR	Yes (17 cycles)
15	AML	2940	PD	No
16	AML	2940	PD	No
17	Myeloma	3000 over 1hr	PD	No
18	NHL	3000 over 1hr	NA	No
19	MDS (RAEB-1)	3000 over 1hr	NA (PD)	No
20	CTCL	2940->2205	PR	Yes (13 cycles)
21	AML	2940	SD	Yes (4 cycles)
22	NHL (DLBCL)	2940	PD	No
23	NHL (DLBCL)	3780	PD	No
24	AML	3780	PD	No
25	AML	3780	PD	Yes (1.5 cycles)
26	AML	3780	NA	No

NA=not assessable, SD=Stable Disease, CR=Complete Remission, MLFS= Morphologically Leukemia Free State, PD=Progressive Disease

Conclusions

CPI-613 is a first in class non-redox active lipoate derivative that has completed a phase I clinical trial for patients with relapsed or refractory hematological malignancies.

❑ MTD identified at a dose of 2940 mg/m² with no marrow suppression seen.

❑ Of the 21 patients evaluable, eight achieved a response of stable disease or better for a response rate of 38%.

❑ All 3 MDS patients who were evaluable for response achieved a response of stable disease or better

❑ A second phase I trial in combination with cytarabine and mitoxantrone is ongoing for relapsed AML patients and a single agent phase II trial in MDS is under review.

We gratefully acknowledge support from: Cornerstone Pharmaceuticals, The Doug Coley Foundation for Leukemia Research, The MacKay Foundation for Cancer Research, and the Frances P Tutwiler Fund. TSP is supported by NCI 1K08CA169809-01.