Phase I study of CPI-613 in combination with Bendamustine for relapsed and refractory T cell lymphoma

Wake Forest School of Medicine

Zanetta S. Lamar[1], Rakhee Vaidya[1], Scott Isom[2], Zachariah McIver[1], Anne Beaven[3]

[1] Department of Hematology and Oncology, [2] Department of Biostatistics, Wake Forest Comprehensive Cancer Center

[3] Department of Hematology and Oncology, Duke Cancer Institute

Background

CPI-613 is a lipoate derivative that has shown activity in hematologic malignancies. CPI-613 selectively targets the pyruvate dehydrogenase complex (PDC) and α-ketoglutarate dehydrogenase complex (KGDHC) in tumor cells and inhibits the catalytic and regulatory functions of the PDC and the KGDHC causing alterations of mitochondrial enzyme complex activities and altering redox status, leading to apoptosis, necrosis and autophagy of tumor cells. The anti-tumor activity of CPI-613 is evident in various cancer cell lines, xenograft animal tumor models and clinical trials against a diverse group of cancers. Patients tolerate CPI-613 as a single agent at doses up to 3,000 mg/m^2, according to Phase 1 trials in patients with solid tumors and hematologic malignancies. Bendamustine has shown single agent activity in the relapsed lymphoma setting with response rates of approximately 50% for T cell NHL.

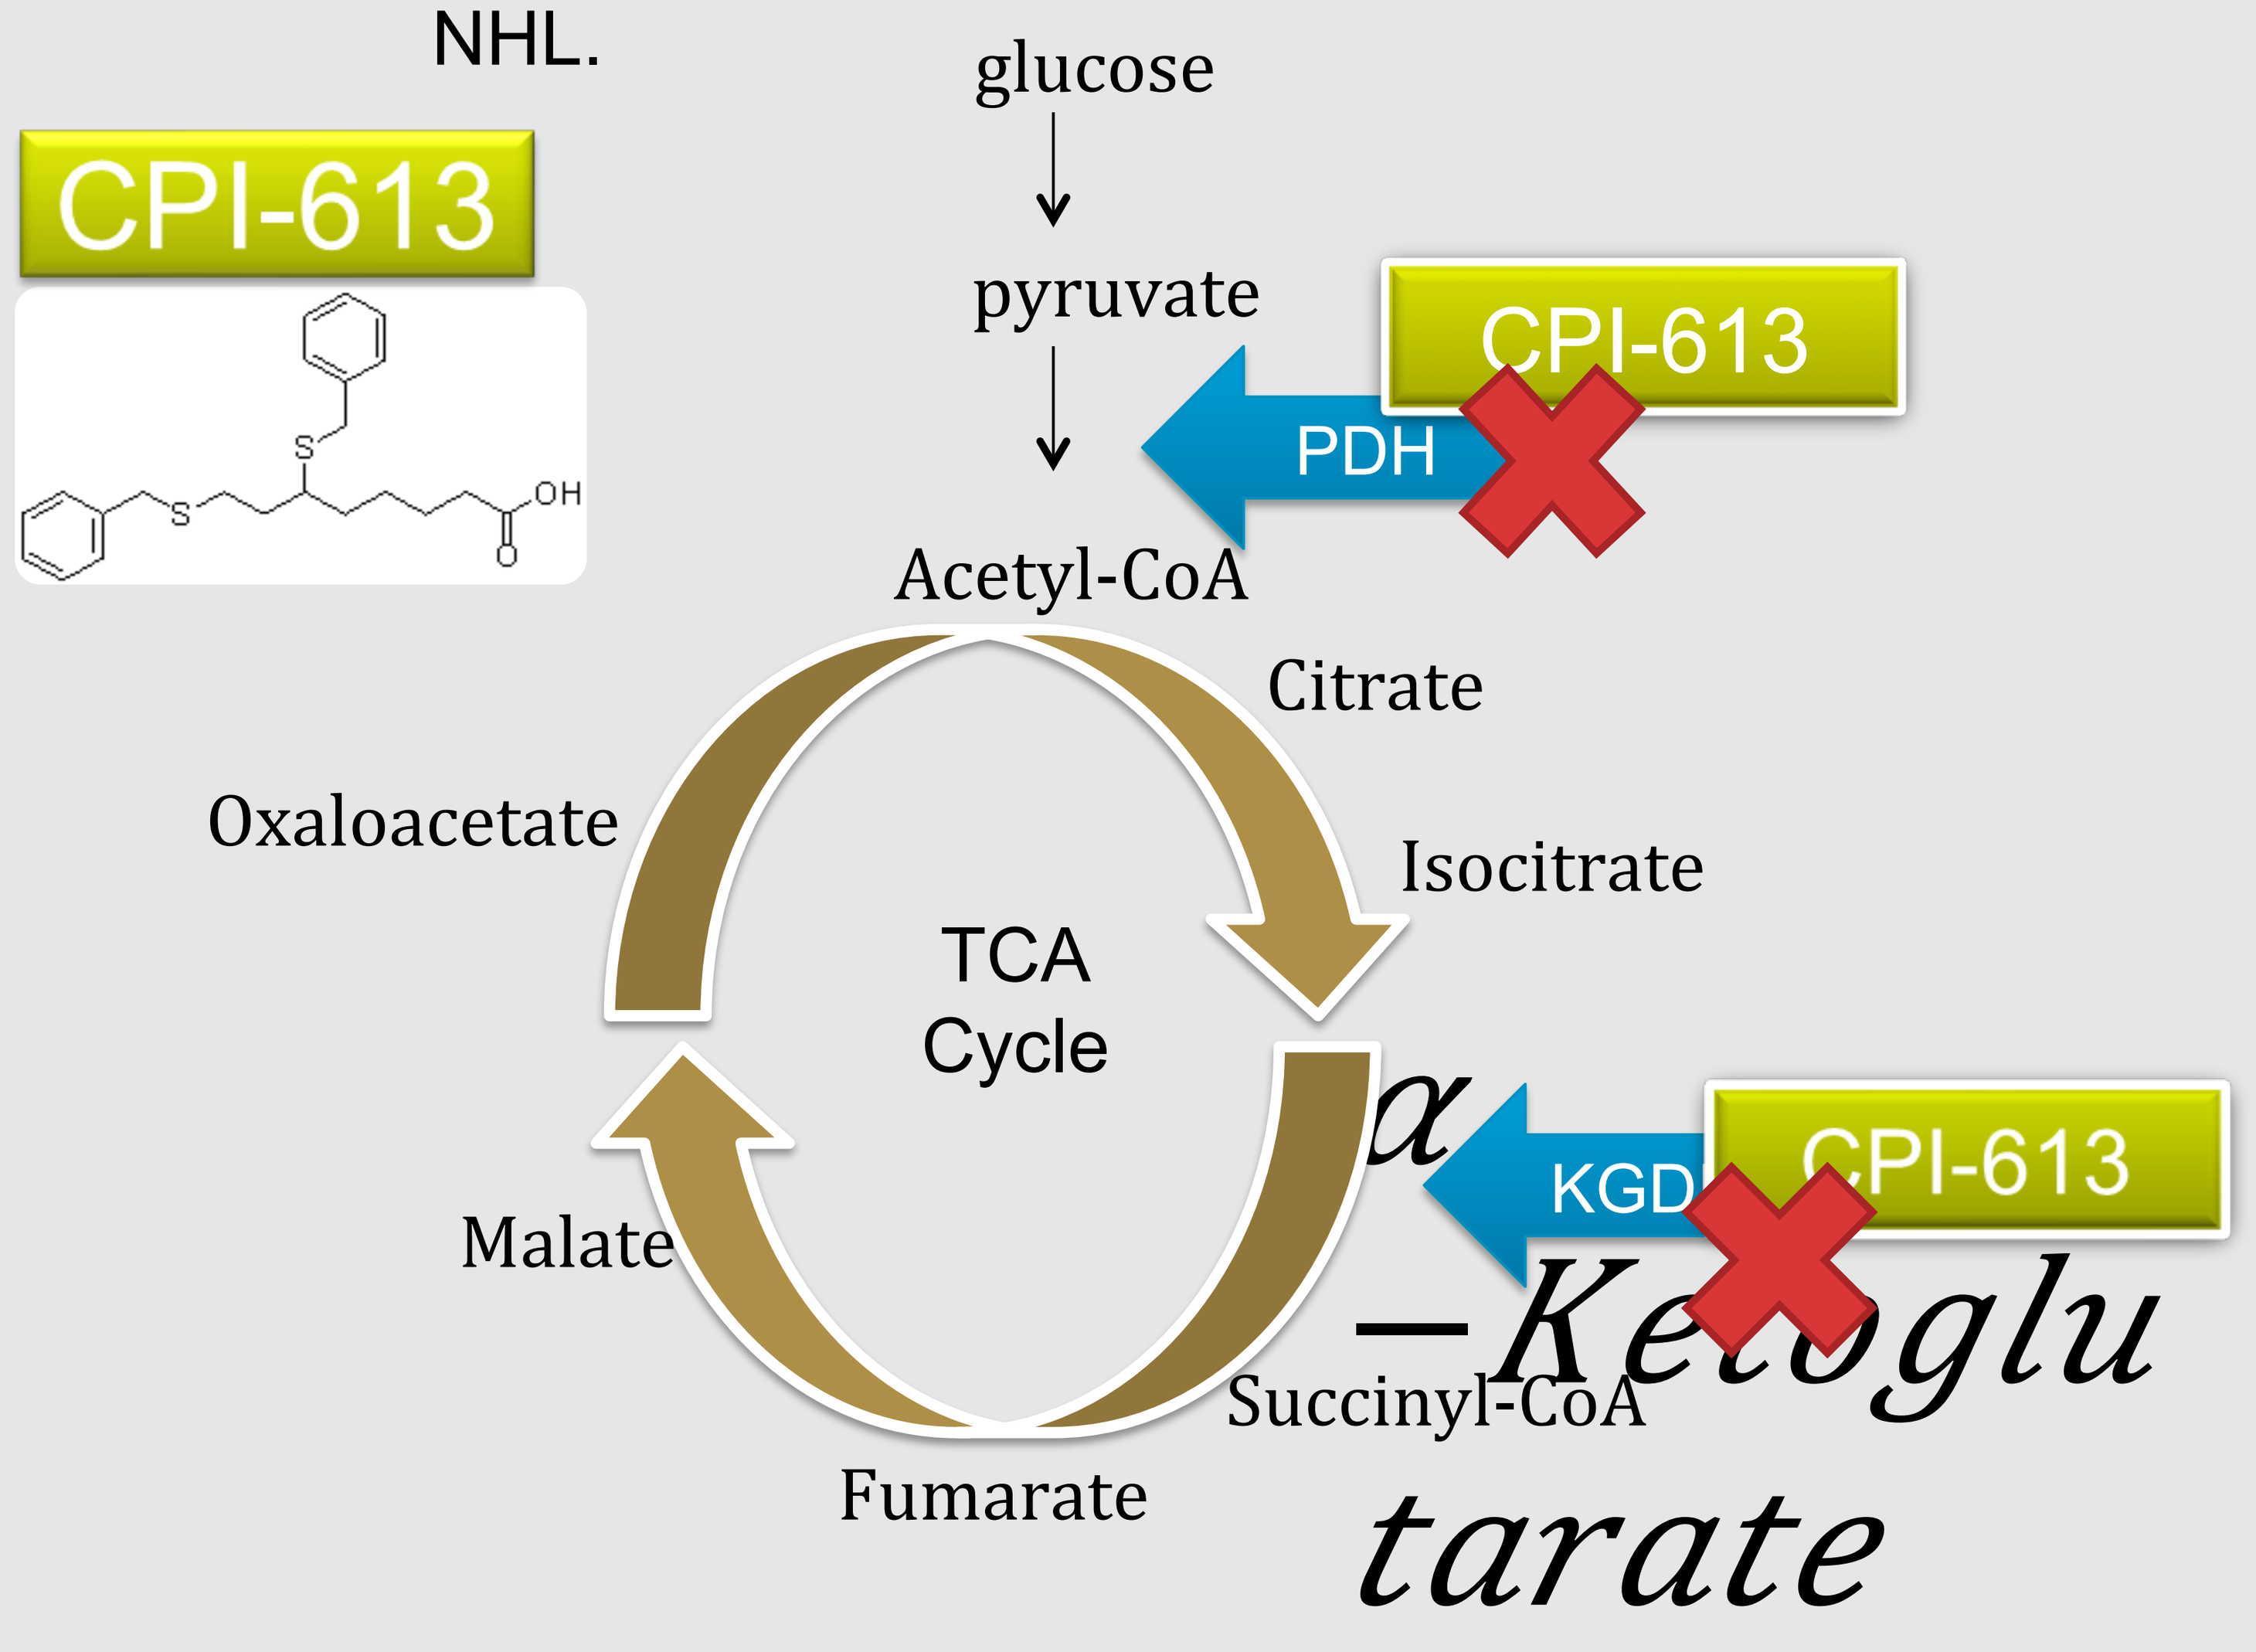
CPI-613
OH
glucose
pyruvate
CPI-613
PDH
Acetyl-CoA
Citrate
Isocitrate
Oxaloacetate
TCA Cycle
Malate
Fumarate
Succinyl-CoA
KGD
CPI-613
α −Ketoglu tarate

Introduction

This is a Phase I study assessing the safety and efficacy of escalating doses CPI-613 in combination with Bendamustine in patients with relapsed or refractory T-cell NHL. Our goal is to determine the Maximum Tolerated Dose (MTD) of CPI-613 when used in conjunction with Bendamustine in patients with relapsed or refractory T-cell NHL, who have or have not received hematopoietic cell transplant.

Methods

Open label, modified 3+3 dose escalation clinical trial evaluating CPI-613 and Bendamustine combination therapy. CPI-613 was given at escalating doses starting at 2,000mg/m^2 over 2 hrs on days 1-4, and on days 8, 11, 15 and 18. Bendamustine was infused at 90 mg/m^2 on days 4 and 5 of each treatment cycle.as shown below. Each treatment cycle was 4 weeks and repeated up to six cycles. There was no intra-patient dose escalation.

Study Methodology

Day	1	2	3	4	5
WEEK (4 week cycle)	CPI	CPI	CPI	CPI Bendamustine	Bendamustine
	CPI			CPI	
	CPI			CPI	
REST					

Results

As of July 27, 2016, eight subjects have received at least one dose of study drug. Eight patients are evaluable for safety and five patients are evaluable for response. The most common grade 3 or higher toxicities were lymphopenia and neutropenia and occurred in 4 subjects. A patient dosed at 2750 mg/m^2 had a dose limiting toxicity of grade 3 acute kidney injury and grade 4 lactic acidosis. The protocol was later amended to discontinue dose escalation at doses of 2750 mg/m^2 or higher and to expand the 2500mg/m^2 cohort

Table 2: Treatment Group (n=7, 5 evaluable)

	CR*	PR	POD
N	3	1	1
%	60%	20%	20%
ORR	80%		

Conclusions

This first reported study of CPI-613 administration in combination with Bendamustine in subjects with relapsed or refractory T cell lymphoma showed a good safety profile and an excellent overall response rate of 80% with CRs in all three patients with peripheral T cell lymphoma, NOS. Although numbers are small, continued investigation is warranted as these response rates in a poor risk population of patients with relapsed/refractory T cell lymphoma are very exciting

Table 3: Responder Data

Pt#	Treatment Cycles (Completed)	Time to Response (Weeks)	Duration of Response (Months)
1	5	9	3.5
4	3.5	10	Not Reached (NR)
6	6	7	NR

Clinical trial: NCT02168907. Research and Biostatistics Shared Resource services were supported by the Wake Forest Baptist Comprehensive Cancer Center's NCI Cancer Center Support Grant P30CA012197.

References

1. Lee KC, Maturo C, Perera CN, Luddy J, Rodriguez R, Shorr R. Translational assessment of mitochondrial dysfunction of pancreatic cancer from in vitro gene microarray and animal efficacy studies, to early clinical studies, via the novel tumor-specific anti-mitochondrial agent, CPI-613. Ann Transl Med. 2014;2: 91.
2. Pardee TS, Lee K, Luddy J, et al. A phase I study of the first-in-class antimitochondrial metabolism agent, CPI-613, in patients with advanced hematologic malignancies. Clin Cancer Res. 2014;20: 5255-5264.
3. Damaj G, Gressin R, Bouabdallah K, et al. Results from a prospective, open-label, phase II trial of bendamustine in refractory or relapsed T-cell lymphomas: the BENTLY trial. J Clin Oncol. 2013;31: 104-110.