A Phase II Clinical Trial of CPI-613® (devimistat) in Patients with Relapsed or Refractory Burkitt Lymphoma/Leukemia or High-grade B-cell Lymphoma with Rearrangements of MYC and BCL2 and/or BCL6 (DHL/THL)

Memorial Sloan Kettering Cancer Center

Ariela Noy,[1] Timothy S. Pardee,[2,6] Liana Nikolenko,[3] Raphael E. Steiner,[4] Jeremy Abramson,[5] Kieron Dunleavy, and Sanjeev Luther.[6]

[1]Memorial Sloan Kettering Cancer Center and Weill Cornell Medical College,NY; [2]Wake Forest Baptist Comprehensive Cancer Center, Winston-Salem, [3]City of Hope, Duarte, CA; [4]MD Anderson, Houston, TX [5]George Washington University Cancer Center, Washington, DC and [6]Rafael Pharmaceuticals, Cranbury, NJ.

BACKGROUND

CPI-613® (devimistat): Devimistat is an investigational new drug for the treatment of certain cancers. Devimistat is a non-redox active analogue of lipoic acid, a required cofactor for key mitochondrial enzymes of the Tricarboxylic Acid (TCA) cycle including pyruvate dehydrogenase (PDH) and ketoglutarate dehydrogenase (KGDH)

Rationale for targeting mitochondrial metabolism in Burkitt Lymphoma and DHL/THL:

- Burkitt Lymphoma (BL) is a highly aggressive hematologic B-cell malignancy classically characterized by the overexpression of c-Myc. Similarly, highly aggressive subset of diffuse large B cell lymphoma, DHL/THL, is also driven by the Myc oncogene
- Molecular characterization of c-Myc-translocation in cancer has demonstrated a key link to altered metabolism including mitochondrial metabolism
- Devimistat is a dual inhibitor of PDH and KGDH specific to cancer cells. Devimistat selectively targets the altered form of mitochondrial metabolism in tumor cells, causing changes in mitochondrial enzyme activities and redox status, leading to apoptosis, necrosis, and autophagy of tumor cells
- In a preclinical model system when leukemia cells were forced to overexpress c-Myc, they became more sensitive to devimistat mediated cell killing. Devimistat inhibited growth of two human cell lines derived from Burkitt lymphoma patients (DG75 and Raji)
- Clinical Experience in Burkitt Lymphoma: In a Phase I study for relapsed or refractory hematologic malignancies at one patient on this study diagnosed with multiply relapsed Burkitt lymphoma demonstrated a definitive clinical response to the treatment sustained for one year followed by complete resection
- Phase II trial of devimistat monotherapy is ongoing in patients with relapsed or refractory Burkitt lymphoma/leukemia or high-grade B-cell lymphoma with rearrangements of MYC and BCL2 and/or BCL6. This multicenter trial is sponsored by Memorial Sloan Kettering Cancer Center

MECHANISM OF ACTION

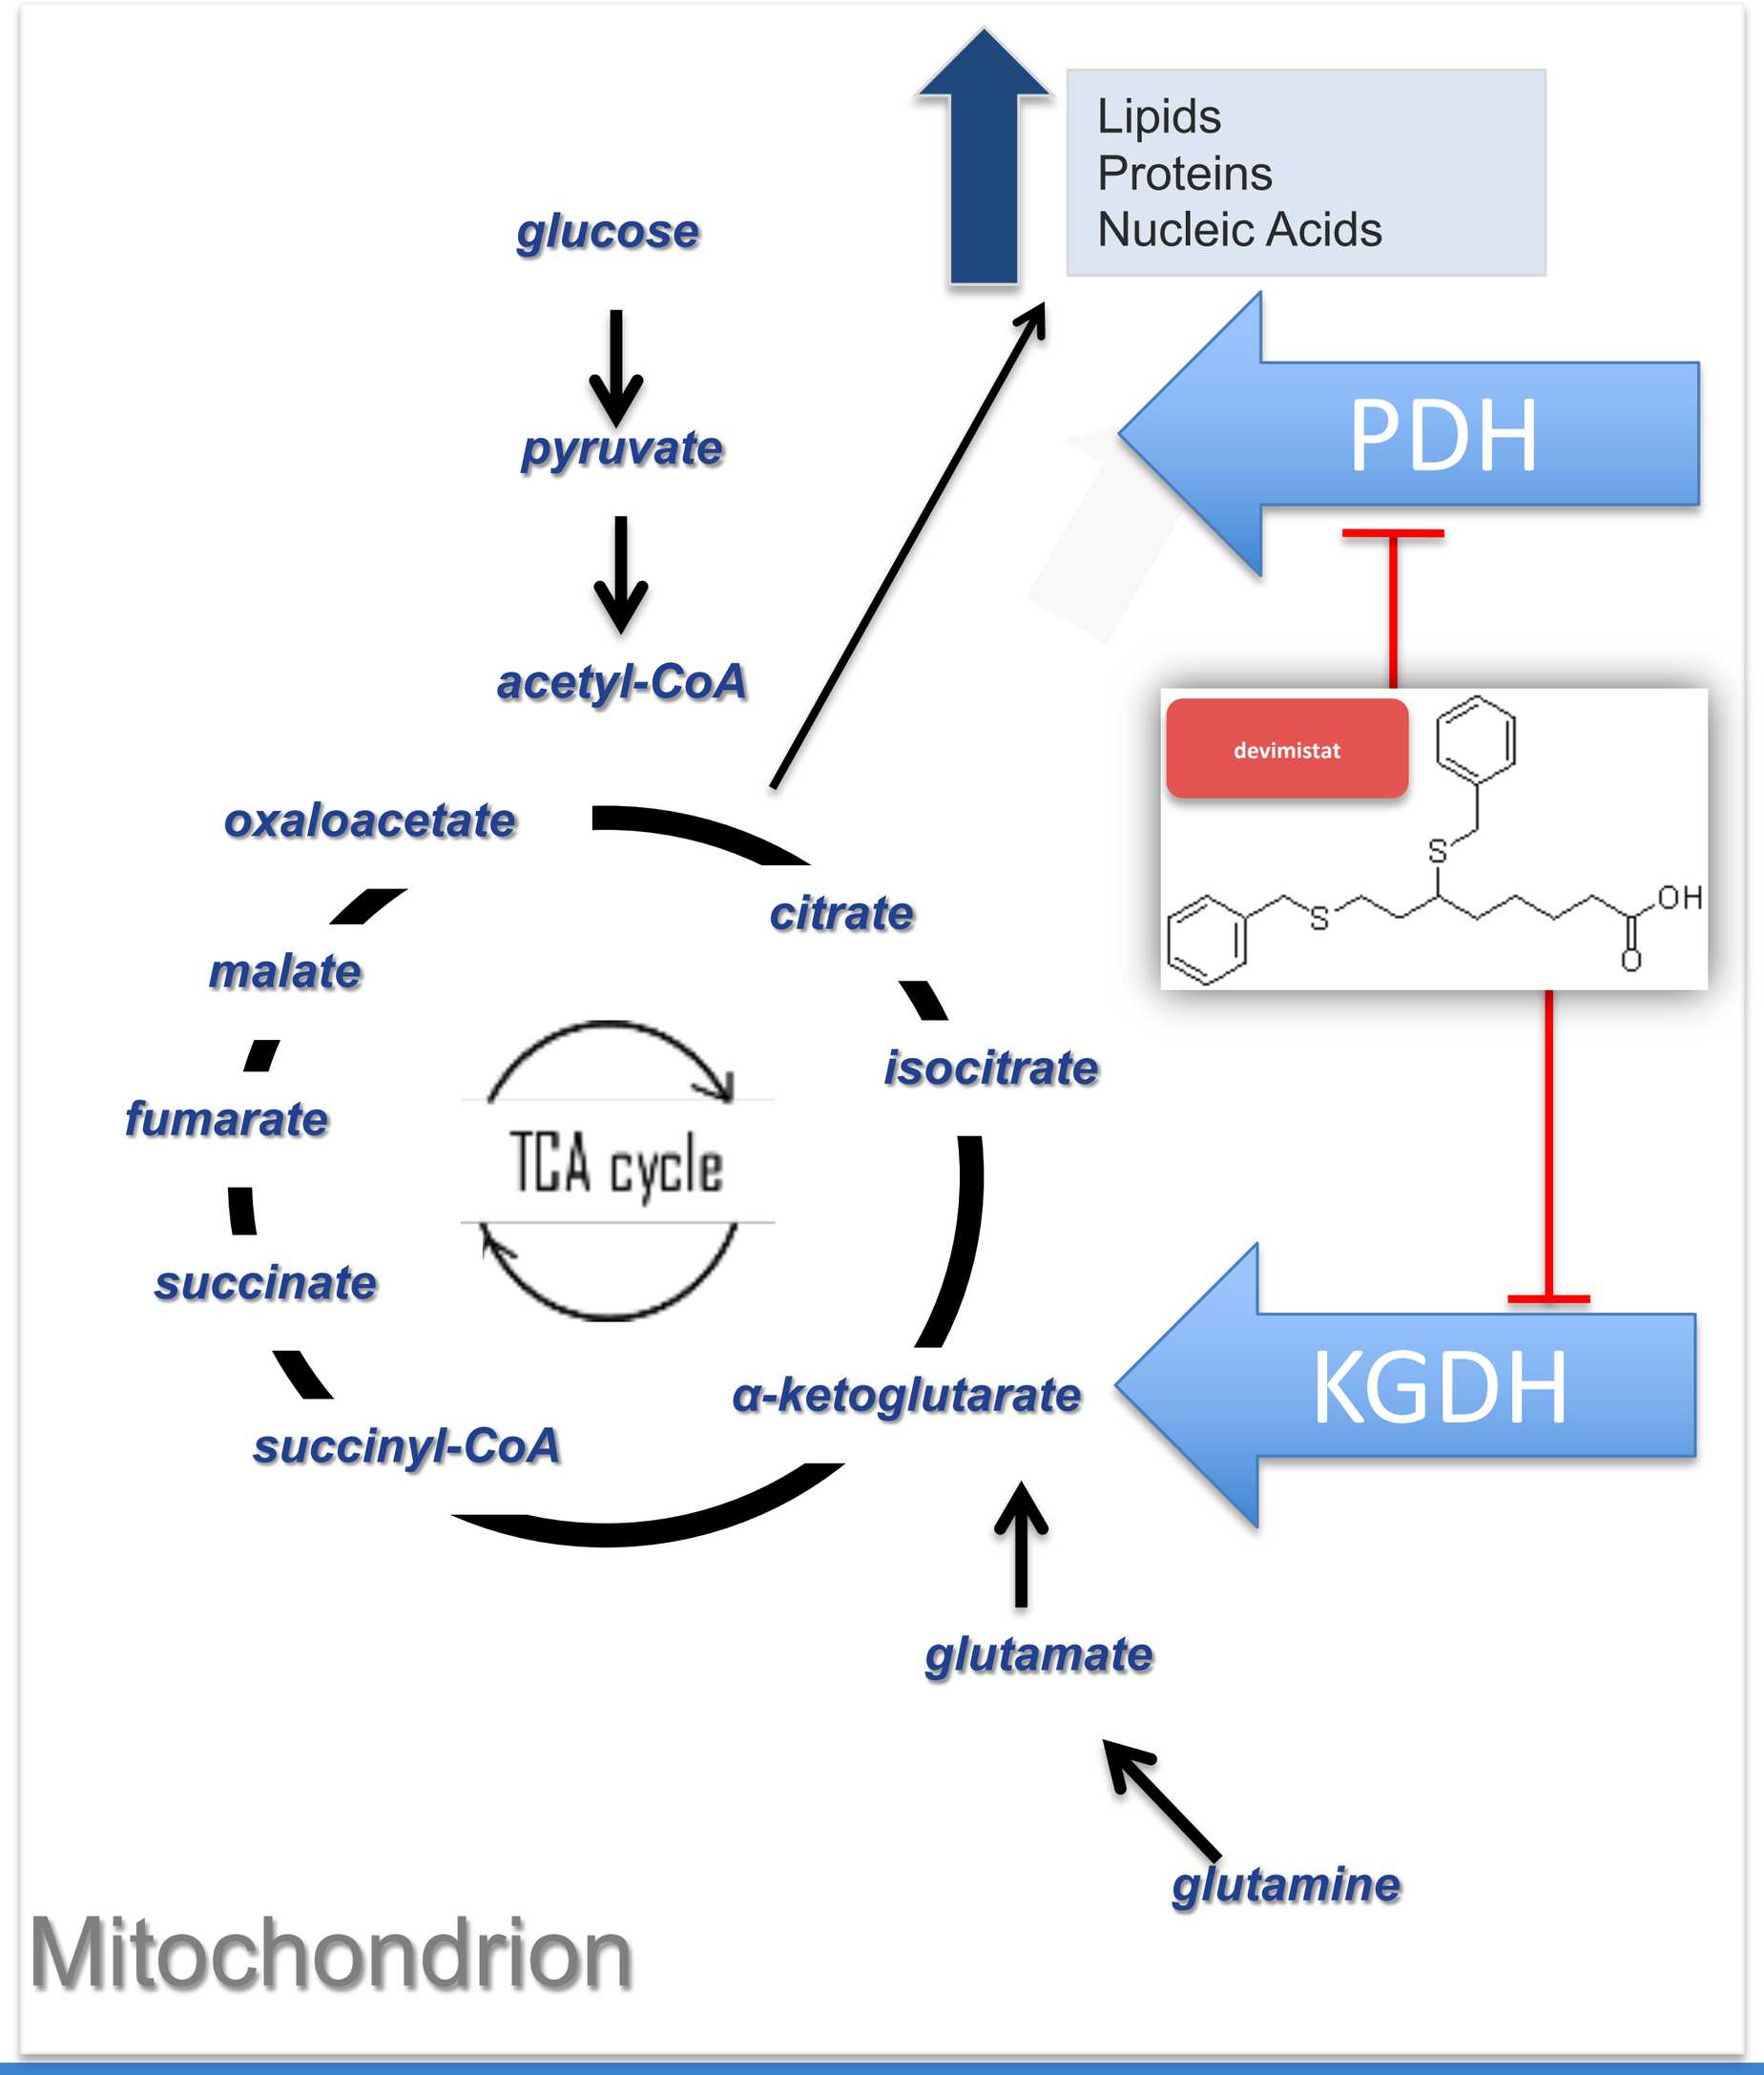
Lipids
Proteins
Nucleic Acids
glucose
pyruvate
acetyl-CoA
PDH
devimistat
oxaloacetate
citrate
malate
isocitrate
fumarate
TCA cycle
succinate
α-ketoglutarate
KGDH
succinyl-CoA
glutamate
glutamine
Mitochondrion

Cancer cells extensively reconfigure normal metabolism:

- ❖ re-regulated carbon flux for controlled provision of metabolic building blocks needed for growth and increased requirement for carbon backbones

Two major re-regulated control points input virtually all carbon into cancer cell Tricarboxylic Acid (TCA) cycle:

- ❖ Pyruvate Dehydrogenase (PDH); inputs pyruvate carbon
- ❖ Alpha-Ketoglutarate Dehydrogenase (KGDH); inputs glutamine-derived carbon

Multi Targeted Approach – selectively blocks PDH and KGDH, through their tumor cell re-regulation, each by a distinct mechanism, ultimately triggering cell death, while sensitizing to most other drugs.

STUDY OBJECTIVES, STUDY DESIGN & LOCATION

Primary Objective:

- To determine the overall response rate of devimistat in patients with relapsed or refractory Burkitt Lymphoma (BL) and double hit diffuse large B cell lymphoma (DHL), analyzed separately

Secondary Objectives:

- To evaluate the duration of response, progression-free survival (PFS), overall survival (OS), and assess the safety of devimistat in patients with relapsed or refractory BL and double hit DHL, analyzed separately

Exploratory Objectives:

- To correlate primary and secondary outcomes with pre-treatment biomarkers including variances in immunohistochemistry and pretreatment cytokine profiles
- Specimens collected during baseline and on treatment will also be used for unspecified future research

STUDY DESIGN: This is an open-label multicenter, single-arm study

LOCATION: United States

KEY ELIGIBILITY CRITERIA

Key Inclusion Criteria:

1. Age ≥ 18 years with Burkitt Lymphoma or high-grade B-cell lymphoma with rearrangements of MYC and BCL2 and/or BCL6 (DHL/THL) confirmed at enrolling institution
2. Failure of at least one previous line of therapy
3. ECOG PS ≤ 3;
4. Failure after prior bone marrow transplant, or ineligible for or opted not to participate in bone marrow transplantation for Burkitt Lymphoma, or DHL/THL
5. Measurable disease as defined RECIL criteria (2017) or isolated bone marrow involvement
6. Creatinine Creatinine clearance <=40cc min
7. AST and AL≤ 5x upper normal limit (ULN)
8. Total bilirubin ≤1.5x ULN (unless related to hemolysis, Gilbert's syndrome, or involvement by lymphoma; if involvement by lymphoma: total bilirubin ≤ 3.0x ULN)

Key Exclusion Criteria:

1. A chemotherapy regimen with stem cell support within 3 months
2. Any clinically unstable medical condition despite present therapy (e.g. uncontrolled infection)
3. Platelets < 50,000/mm3 unless attributable to marrow involvement
4. Serious medical comorbidity increasing risk for toxicity
5. Active CNS parenchymal disease. Leptomeningeal disease is allowed as long as the CSF has cleared for more than 4 weeks and the patient is receiving maintenance intrathecal/intra Ommaya therapy
6. Any active uncontrolled bleeding or bleeding diathesis

STUDY DESIGN SCHEMA

Diagnosis: Pathology review or biopsy for:
Relapsed or Refractory Burkitt Lymphoma/Leukemia
[COHORT 1, N=17]
OR
High-grade B-cell lymphoma with rearrangements of MYC and BCL2 and/or BCL6 [COHORT 2, N=17]

↓

Informed Consent

↓

Induction Treatment:
devimistat [2,500 mg/m^2/day IV] over 2 hours on Days 1-5 for Cycle 1 & 2
(Each Cycle is 14 days)

↓

Maintenance Treatment:
devimistat [2,500 mg/m^2/day IV] over 2 hours on Days 1-5 for all Cycles thereafter
(Each Cycle is 21 days)

TIME TO COMPLETION

- The study is currently recruiting patients, with targeted enrollment of total 34 subjects
- Accrual is expected to be 10 patients/year for up to 3 years and one year of follow up for data analysis
- The study duration will be 4 years with expected completion date of December 2021
- Patients will continue to be followed for progression and survival until they complete or withdraw from treatment.
- Treatment can be used as a bridge to transplant.

REGISTRATION:

- ClinicalTrials.gov Identifier: NCT03793140

CONTACT INFORMATION:

- For more information on qualification and enrollment, please contact Ariela Noy, MD (noya@mskcc.org) #646=608-3727